Investigator Blinded Randomized Study Evaluating HOCl in the Treatment of Atopic Dermatitis-Associated Pruritus

Brian Berman, MD, PhD Mark Nestor, MD, PhD

Center for Clinical and Cosmetic Research and University of Miami Miller School of Medicine

Introduction

Atopic dermatitis (AD) is a chronic inflammatory skin disease characterized by pruritus. It affects up to 20% of children and 3% of adults worldwide, and its prevalence is increasing.[1] The impact of pruritus in AD can range from mildly distressing to completely disabling.[2]

Hypochlorous acid (HOCl) has been identified as a treatment for pruritus.[3] There are 2 mechanisms by which HOCl may reduce pruritus: 1) by its microbicidal qualities, particularly in the case of Staphylococcus aureus; and 2) by its anti-inflammatory qualities, which help reduce the activity of histamine, leukotriene B4, and interleukin-2, all of which contribute to the pathophysiology of itch.[4-7]

Here we present the results of a 3-day study designed to evaluate the effect of HOCl on pruritus in patients with AD.

Material and Methods

The study was conducted according to the protocol and in compliance with Good Clinical Practice (GCP) and other applicable regulatory requirements. This investigator blinded randomized phase 2 72-hour study investigated the antipruritic effect of HOCl with patients diagnosed with AD. Subjects were enrolled into the study if they had AD as defined by the Hanifin criteria and had a score of >2 on an itch severity scale (0-4).

30 subjects were enrolled over the course of the study, with 20 randomized to the treatment group (HOCl), and 10 randomized to the untreated control group. Subjects who were randomized to the treatment group were instructed to use HOCl BID or PRN for 72-hours while recording the date and time of applications in a study diary. Subjects randomized to the untreated group received no treatment and were only instructed to come to follow-up visits for study-specific assessments.

The 3 primary measures used in this study were the Participant Global Assessment (PGA), Investigator Global Assessment (IGA), and VAS itch score.

RESEARCH POSTER PRESENTATION DESIGN © 2015
www.PosterPresentations.com

PGA was defined as the mean of five 5-point ordinal scales (0- none, 4-severe) for subject rated overall irritation, peeling, stinging, burning, and itching. The IGA was defined as the mean of eight 5-point ordinal scales (0-4, none-severe) for erythema, desquamation, lichenification, overall irritation, excoriation, and subject queried stinging, burning and itching. The VAS itch score was defined as the number of millimeters from the left side of a line that indicates their level of itching (0 mm no itching to 154 mm most itching). Along with the 3 primary measures, incidence of unexpected adverse events (AEs) and serious adverse events (SAEs) and incidence of local skin reactions leading to discontinuation were recorded as well. Measurements were taken at baseline, 24-hours, and 72-hours post treatment.

Photographic evidence was used to confirm evaluations.

Results

Thirty subjects were enrolled into the study in a 2:1 ratio (treated:untreated), and 29 were included in the final analysis. The mean VAS itch score between the 2 groups were similar at baseline (Table 1). Mean change in PGA and IGA between baseline and 72-hours were both shown to be significantly different, with a decrease (improvement) in favor of the treatment group (PGA: p-value=0.128; IGA: p-value=0.012) (Figure 1). The mean itch VAS scores between the treated and untreated groups were significantly different between baseline and 72-hours post application, with the percent mean change shown to be significantly lower in the treated group (Figure 2).

At the conclusion of the study, subjects in both groups were separated into those who had less itch (difference in itch between baseline and 72 hours was positive), same itch (difference in itch between baseline and 72 hours was equal to zero), or more itch at 72 hours (difference in itch between baseline and 72 hours was negative) (Figure 3). The analysis showed 73.7% of the subjects in the treated and 30.0% of the subjects in the untreated group experienced a reduction in itching between baseline and 72 hours post application. There were no treatment related discontinuations or SAEs. One subject reported mild to moderate transient facial dryness which spontaneously resolved when that subject applied the gel to the face. Figure 4 shows photographic evidence of a significant decrease in AD characteristics (erythema, dryness, desquamation) at 72 hours.

Table 1. Baseline Investigator Elicited Itch Score (0-4)

Study Group	n	Mean (μ) IGA Itch Score at Baseline	Standard Deviation (σ) of mean
TREATED	20	**1.55**	0.826
UNTREATED	10	**1.70**	0.483

Statistical Test	p-value (α=0.05; β=0.20)	95% Confidence Interval
Independent Samples t-test	**0.601**	-0.732 - 0.432

Figure 1. Mean Change in PGAD3-D1 and IGAD3-D1 With and Without Treatment with HOCl Gel x 3 days

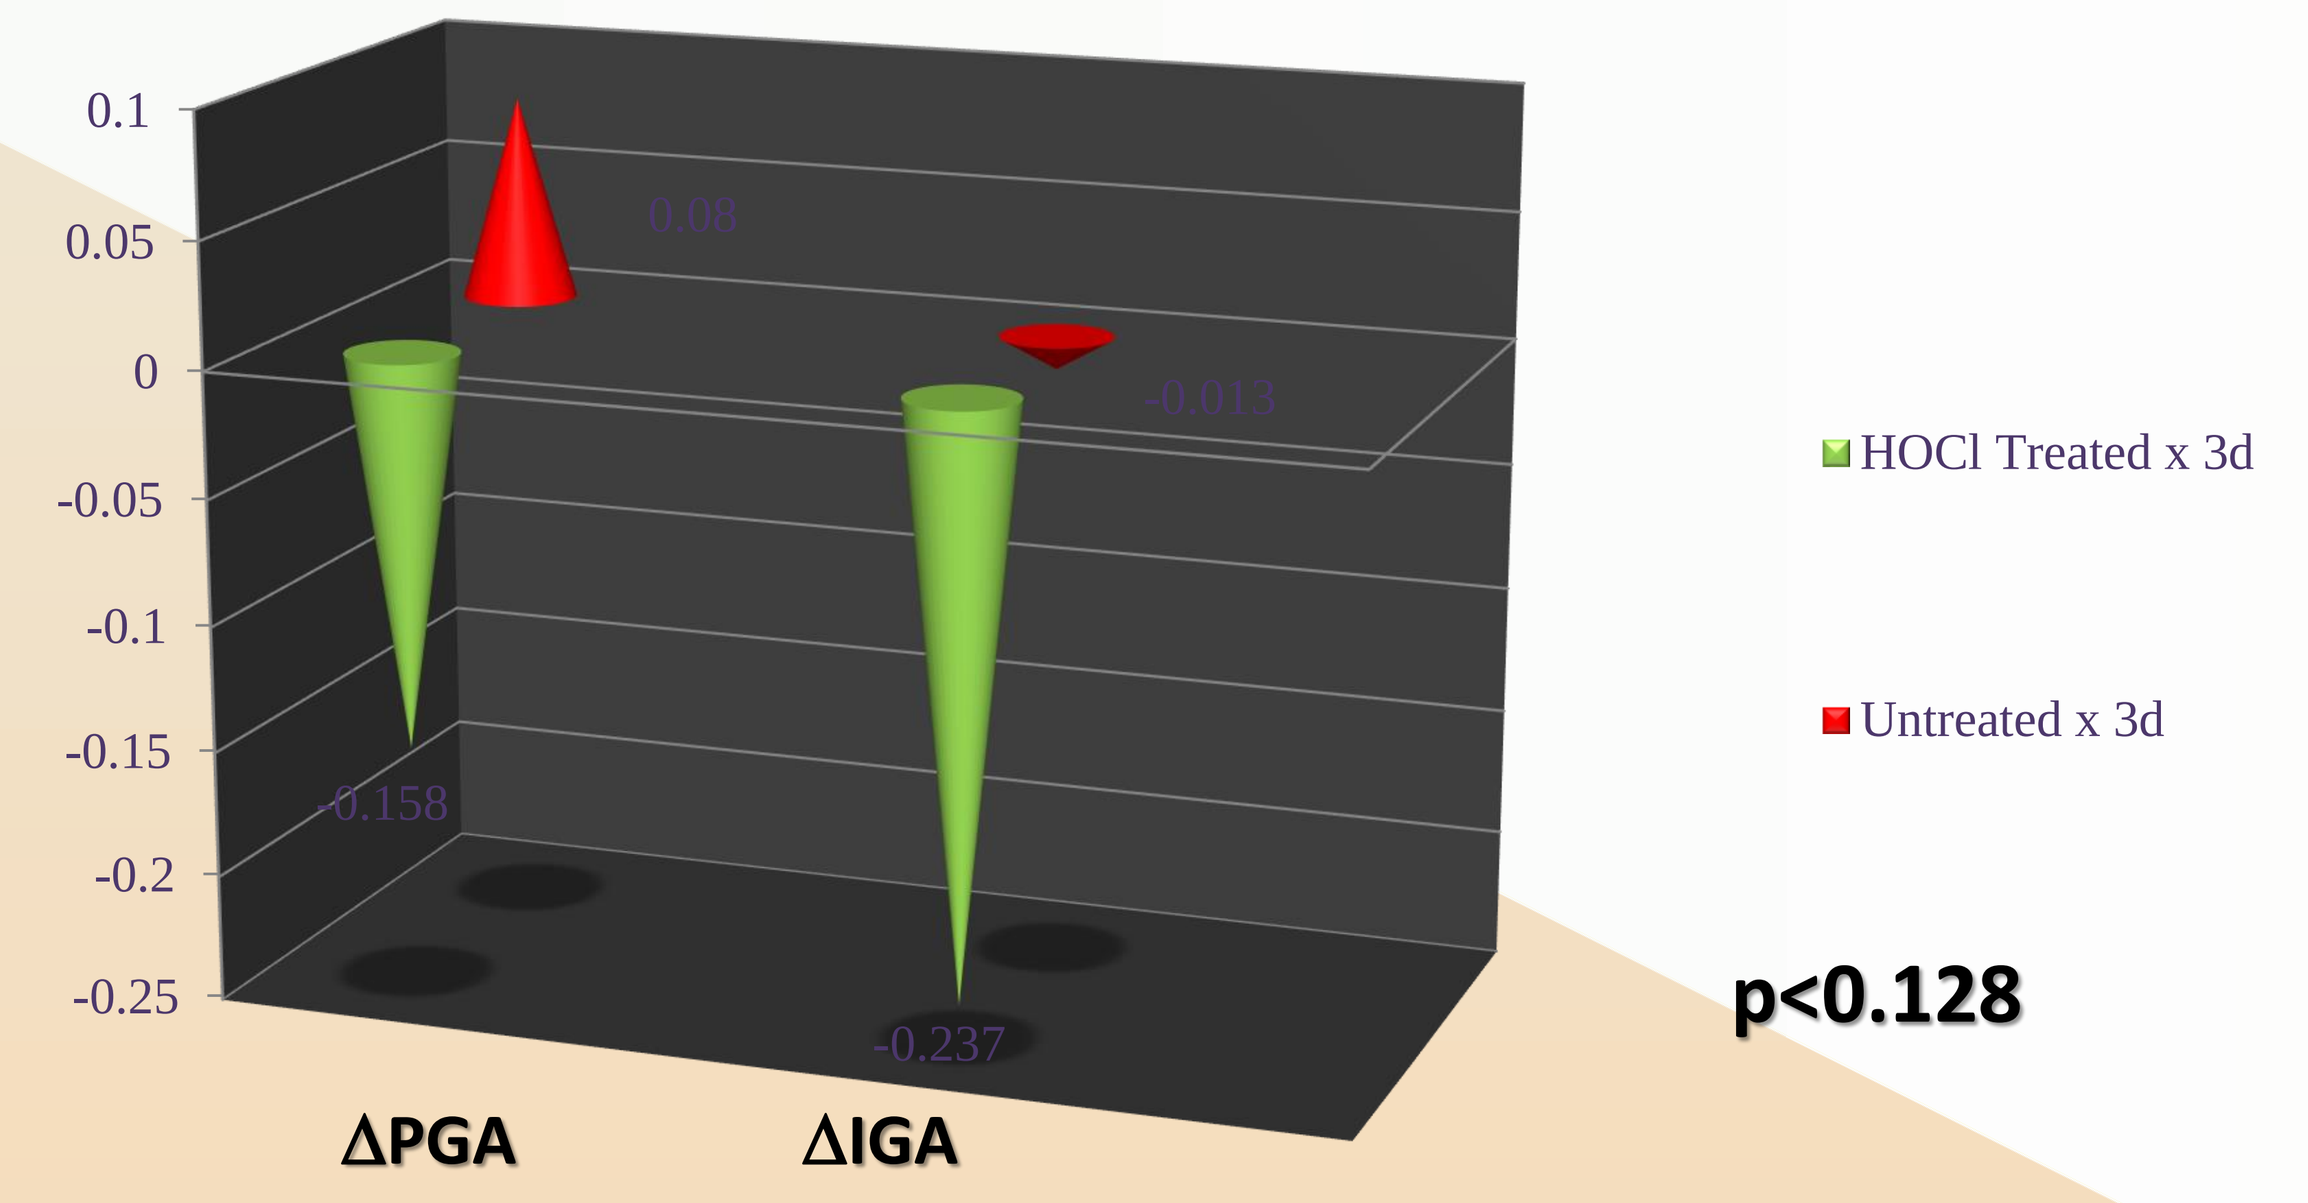
0.1
0.05
0
-0.05
-0.1
-0.15
-0.2
-0.25
0.08
-0.013
-0.158
-0.237
ΔPGA
ΔIGA
HOCl Treated x 3d
Untreated x 3d
p<0.128

Figure 2. Mean % Change in Itch VAS With and Without Treatment with HOCl Gel x 3 days

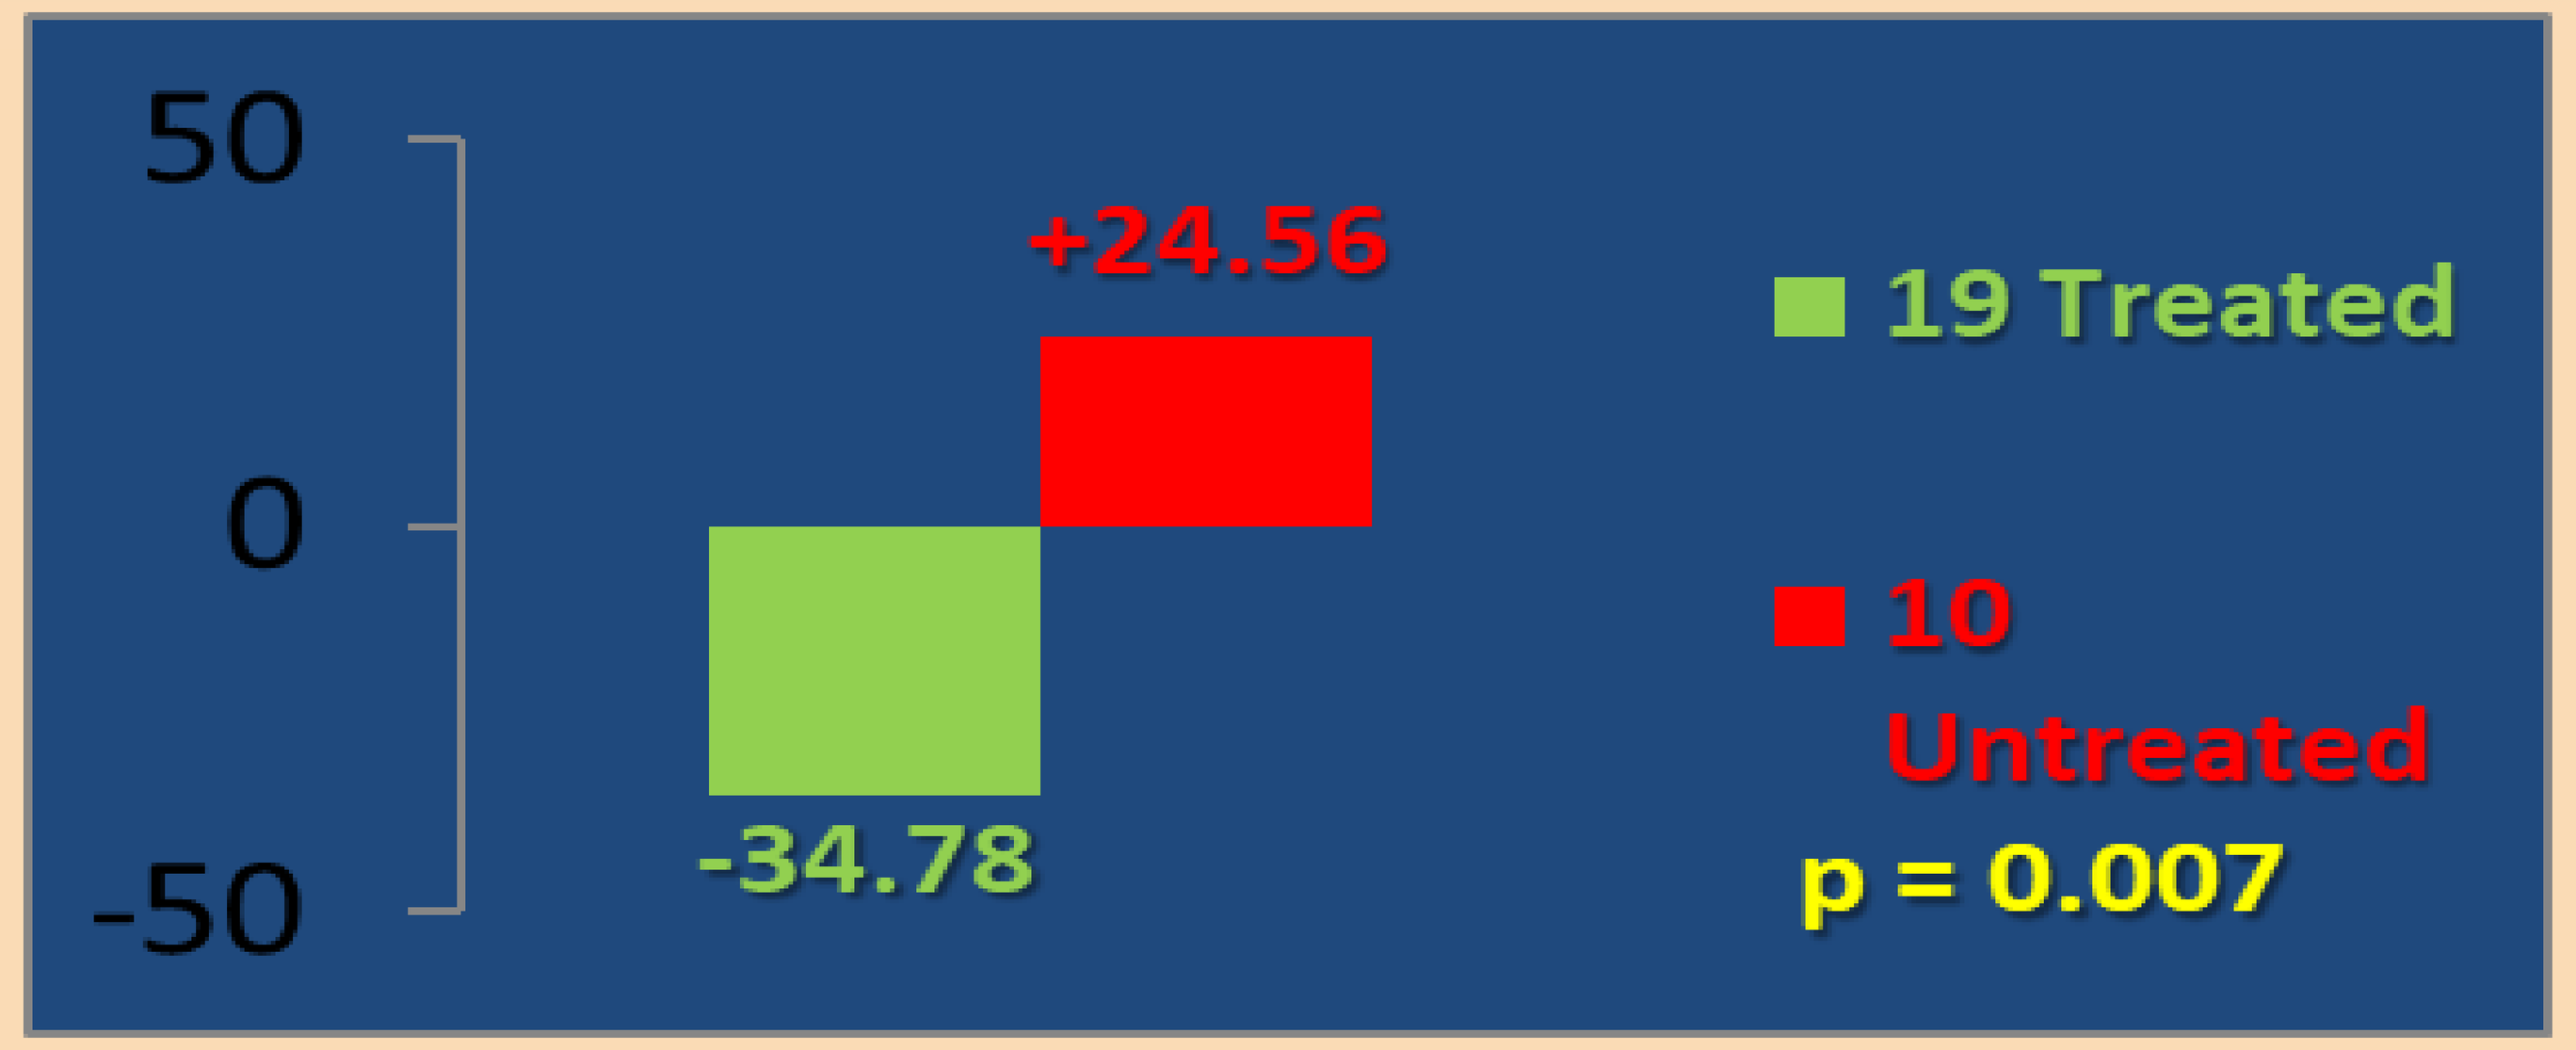
50
0
-50
+24.56
-34.78
19 Treated
10 Untreated
p = 0.007

Figure 3. Effect of with HOCl Gel Treatment on Itch in Atopic Dermatitis - day 3

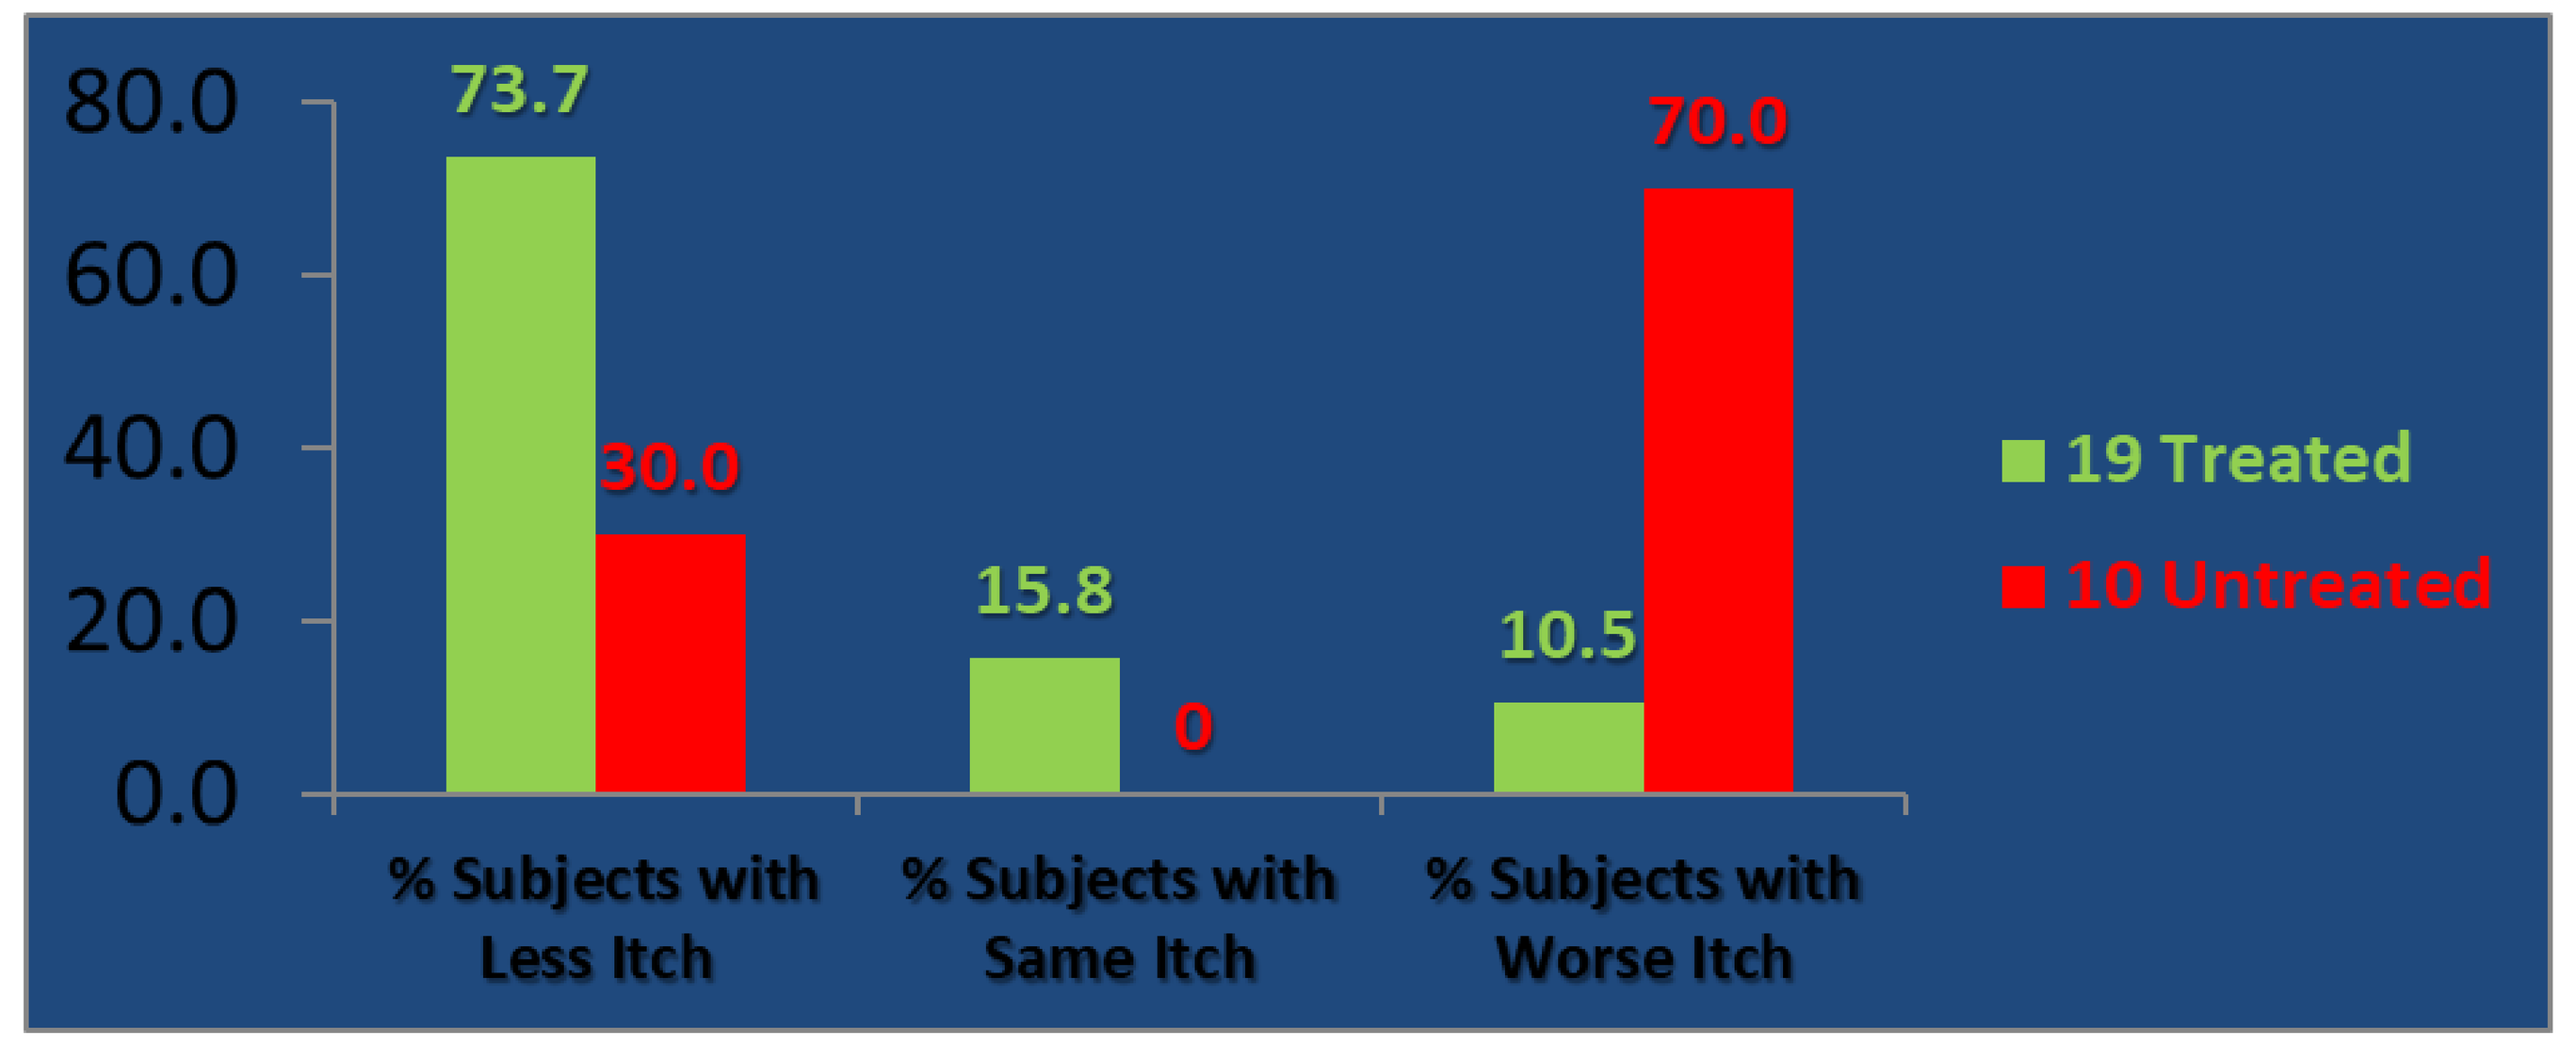
80.0
60.0
40.0
20.0
0.0
73.7
30.0
15.8
0
10.5
70.0
% Subjects with Less Itch
% Subjects with Same Itch
% Subjects with Worse Itch
19 Treated
10 Untreated

Conclusion

This study demonstrated that HOCl leads to a significant reduction in itching associated with AD in as little as 72 hours. Additionally, the twice daily regimen was manageable and easy to follow, as demonstrated by a high rate of compliance.

HOCl is effective over a short period of time with few doses needed. This is a cost efficient and effective method for improving the symptom of pruritus in patients with AD.

Figure 4. Subject treated with HOCl before (left) and 72 hours after (right) treatment

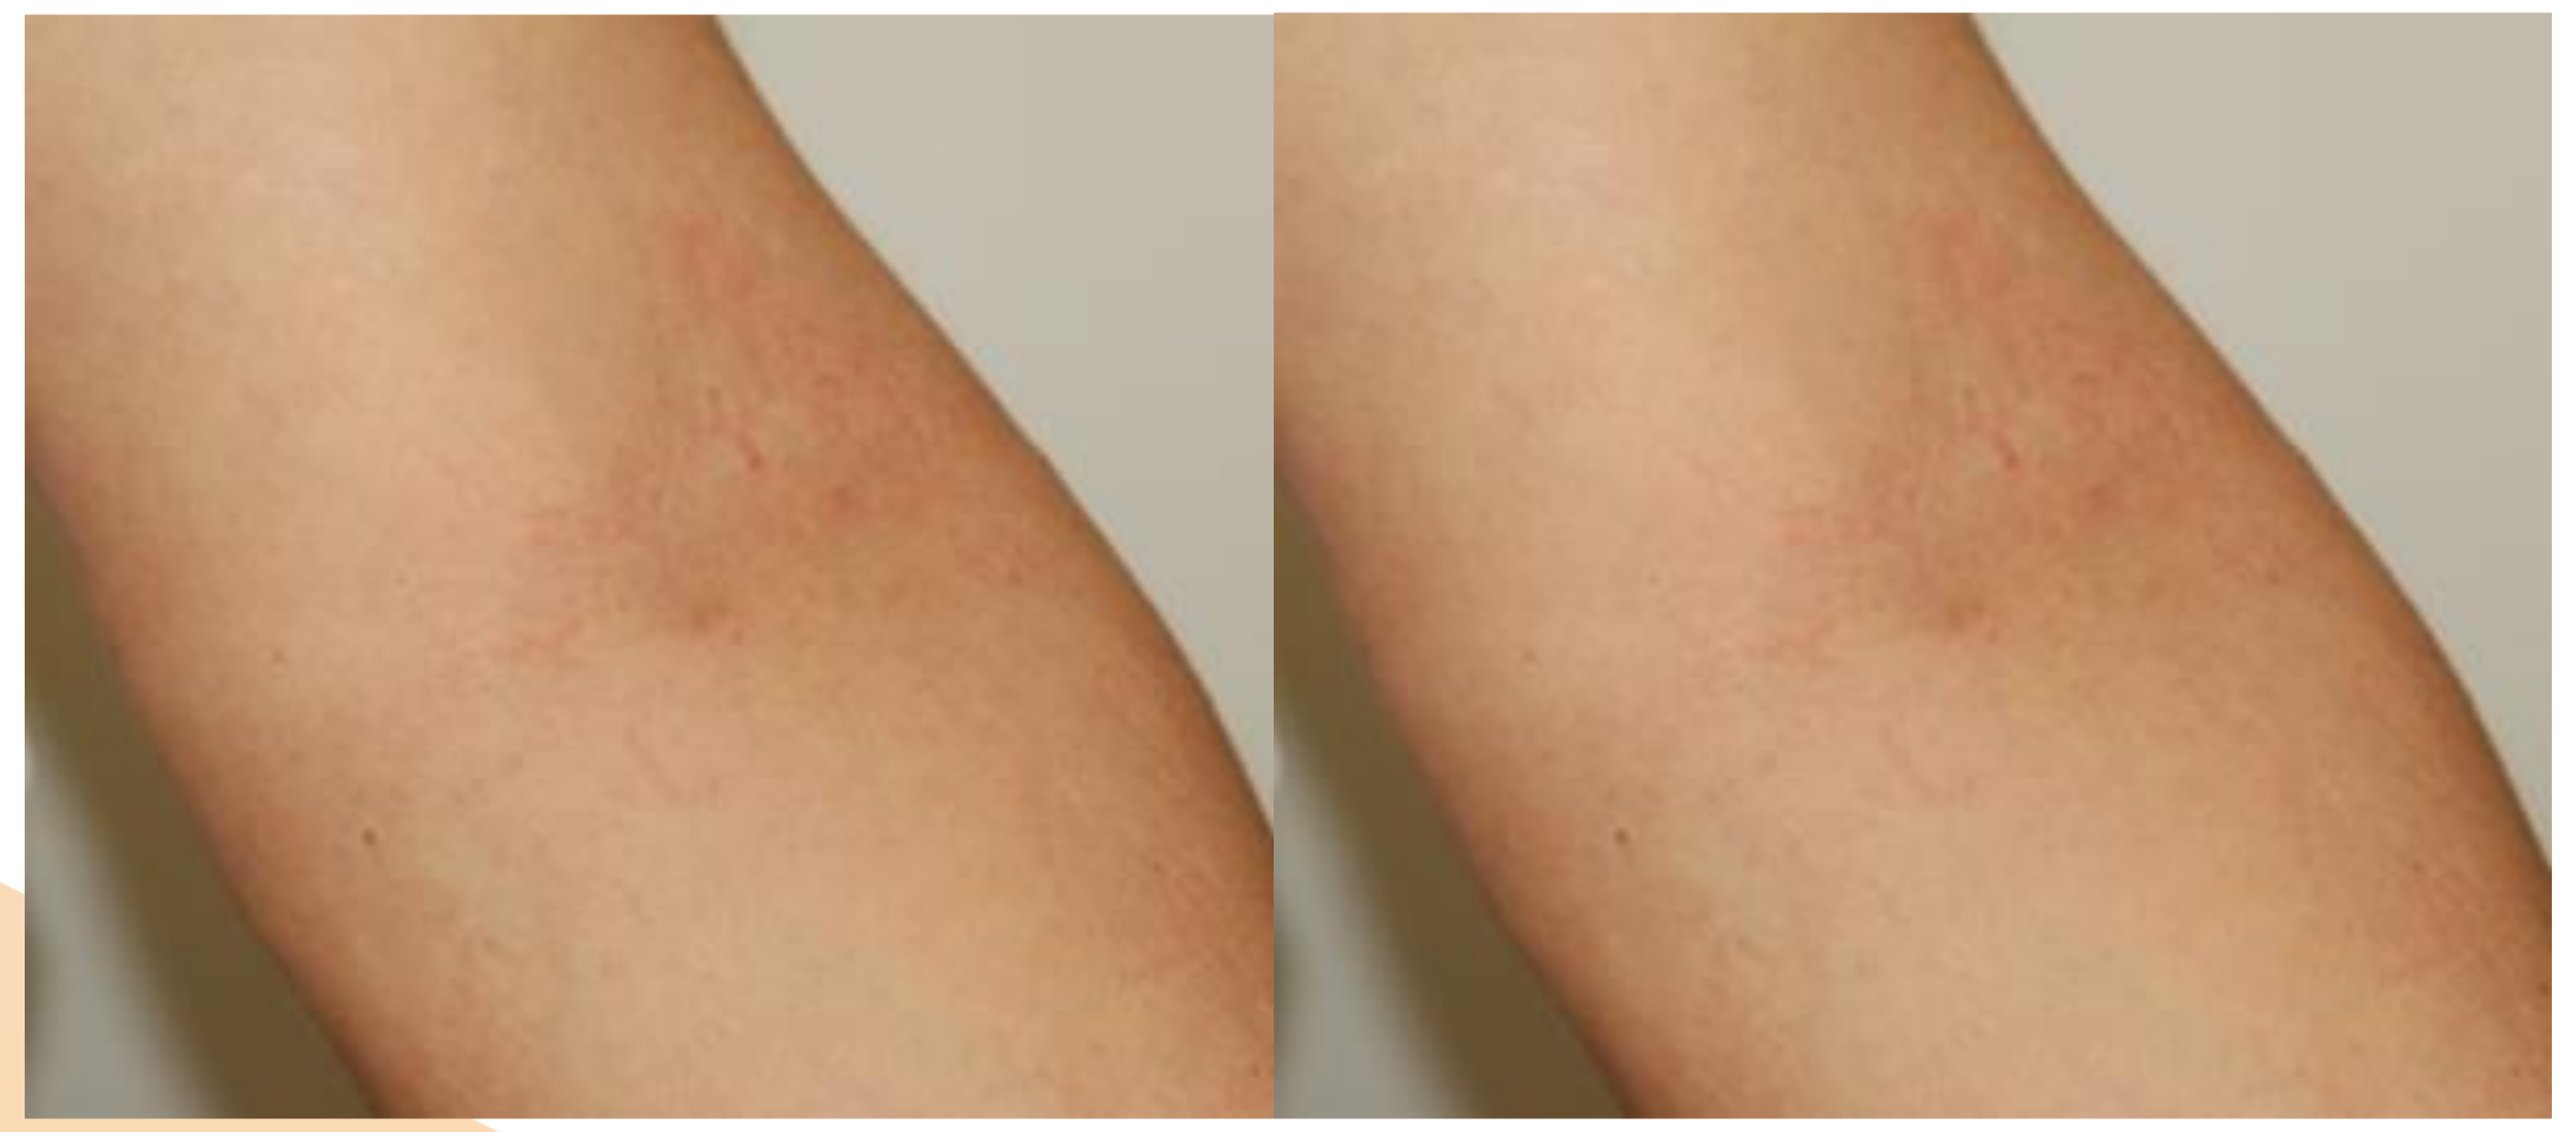

	Day 2	Day 3
Change in VAS score 0-72 hours/Baseline VAS Score x 100%	-59.5%	-84.5%

References

1. Nutten S. Atopic dermatitis: global epidemiology and risk factors. Ann Nutr Metab. 2015;66(suppl 1):8-16.
2. Elmariah SB. Adjunctive management of itch in atopic dermatitis. Derm Clin. 2017;35(3):373-394.
3. Pelgrift RY, Friedman AJ. Topical hypochlorous acid (HOCl) as a potential treatment of pruritus. Curr Derm Rep. 2013; 2:181–190.
4. Garibyan L, Rheingold CG, Lerner EA. Understanding the pathophysiology of itch. Dermatol Ther. 2013;26(2):84–91.
5. Greaves MW. Pathogenesis and treatment of pruritus. Curr Allergy Asthma Rep. 2010;10(4):236–42.
6. Yosipovitch G, Papoiu AD. What causes itch in atopic dermatitis? Curr Allergy Asthma Rep. 2008;8(4):306–11.
7. Friedman AJ, Cash K, Berman B. Hypochlorous Acid is Antiinflammatory and Immunomodulatory. Poster. Presented at the Winter Clinical Dermatology Conference, Kauai, HI, January 18, 2013.

Supported in part by Onset and IntraDerm